Impact of Health Concerns on Speech From Birth to 5 Years

Dawn Savery, MA, CYF-SLP
Tracy Furr, MA, CFY-SLP
University of Central Florida
Department of Communicative Sciences and Disorders

Debra Beckman, MS, CCC-SLP
Beckman and Associates, Inc.

ABSTRACT

Many health concerns can impact speech development, especially during the period of birth to 5 years. How significant are factors such as reflux, oral sensitivity, feeding difficulties, bruxism, drooling and premature birth, on speech development at this early age?

This poster reports the results of statistical analysis of an inclusive retrospective study comparing these factors and speech development for 142 children seen over a 14 month period in a community based therapy clinic.

METHODS

142 Participants

Inclusion Criteria:

- Referral due to developmental concerns
- Age at treatment birth to 5 years
- Background intake form completed between 01/01/06 and 03/01/07

Diagnoses:

- 14 Trisomy 21
- 113 Dysarthria
- 25 Language delay
- 16 Non oral intake
- 22 Speech delay
- 7 Cerebral Palsy
- 5 Hypotonia
- 4 PDD
- 4 Otitis Media
- 25 additional diagnoses

Note of Interest
Of the 142 Participants, 110 had more than 1 diagnosis.

Note of Interest
More than 50% of children with feeding problems had speech difficulties

Procedures

Chart review identifying the following:

- Date of birth
- Prematurity
- Feeding difficulty
- Drooling
- GERD
- Bruxism
- Oral sensitivity
- Speech difficulties

Testing

- Client Background Information Form
- Rosetti Infant Toddler Language Scale - 2005
- Beckman Oral Motor Protocol - 2006
- Photo Articulation Test - 3rd Edition (PAT-3)
- Preschool Language Scale - 4th Edition (PLS-4)

Note of Interest
There was a strong co-occurrence between feeding and speech difficulties

Factors (Total N=142)	Premature		Feeding Difficulties		Drooling		GERD		Bruxism		Oral Sensitivity		Speech Difficulties	
	N=27		N=96		N=33		N=46		N=19		N=58		N=107	
Premature	27/27	100%	25/96	26%	4/33	12%	13/46	28%	2/19	11%	10/58	17%	18/107	17%
Feeding Difficulties	25/27	93%	96/96	100%	28/33	85%	42/46	91%	18/19	95%	10/58	17%	70/107	65%
Drooling	4/27	15%	28/96	29%	33/33	100%	14/46	30%	9/19	47%	24/58	41%	24/107	22%
GERD	13/27	48%	42/96	44%	14/33	42%	46/46	100%	6/19	32%	29/58	50%	31/107	29%
Bruxism	2/27	7%	18/96	19%	9/33	27%	6/46	13%	19/19	100%	17/58	29%	14/107	13%
Oral Sensitivity	10/27	37%	52/96	54%	24/33	73%	29/46	63%	17/19	89%	58/58	100%	41/107	38%
Speech Difficulties	18/27	67%	70/96	73%	24/33	73%	31/46	67%	14/19	74%	41/58	71%	107/107	100%

SUMMARY

This was a large sample size of 142 cases that was inclusive for all referrals to a developmental clinic for a 14 month period. Of all the referrals, 110 had more than one health concern factor.

As detailed in the comparison chart, two factor co-occurrence was greater than 50% when looking at all children with feeding problems who also showed speech problems **73%** (70 our of 96) and of all children who had speech difficulties **65%** (70 out of 107) also had feeding problems.

In comparison with other health concerns,

- 67% of children with prematurity (18 out of 27) had speech difficulties
- 73% of children with drooling (24 out of 33) had speech difficulties
- 67% of children with GERD (31 out of 46) had speech difficulties
- 74% of children with bruxism (14 out of 19) had speech difficulties
- 71% of children with oral sensitivity (41 out of 58) had speech difficulties

IMPLICATIONS

Feeding difficulties and speech difficulties may have a significant co-occurrence.

In general practice more detailed background information is needed during intake regarding early feeding history as well as other health concerns.

In general practice a quantified assessment of oral motor skills is needed to determine if a motor deficit exists, which may impact speech production.

Further research is needed to better define the relationship between feeding difficulties and later speech development.

* Additional information available from info@beckmanoralmotor.com

This study is dedicated to the memory of Evan Ackerbloom